Mouse Pluripotent Stem Cell Motor Neurons Generate Robust Neural Network Activity on Microelectrode Arrays

Steven Stice[1,2] , Anirban Majumder[1] , Brad Culp[1] , Anthony Nicolini[3] , Colin Arrowood[3], Nicholas Solomotis[2] , Raymond Swetenburg[2]

[1] ArunA Biomedical, Inc., Athens, Georgia, United States; [2] Regenerative Bioscience Center, University of Georgia, Athens, Georgia, USA; [3] Axion BioSystems, Atlanta, Georgia, USA

www.arunabiomedical.com

Introduction

Pluripotent stem cell-derived cells that are cost effective and generate robust, uniform results will enhance efforts to identify chemicals that affect the network of activity in the central nervous system (CNS). Currently, rodent neural tissue generates spiking and bursting activity after 7-14 DIV.

- Sourcing rodent tissues can pose logistical drawbacks including sourcing animals and animal prep variability when used in large compound screening activity.
- Quantitatively pre-qualifying each primary tissue isolation based on the cell types is arduous and expensive.
- We have designed a scalable means of generating large numbers of prequalified CNS neurons that can be used in longitudinal microelectrode array (MEA) assays where both phenotype and network activity can be monitored temporally and spatially.

Hb9+ Motor Neurons and glia generated from mouse embryonic stem cell aggregates

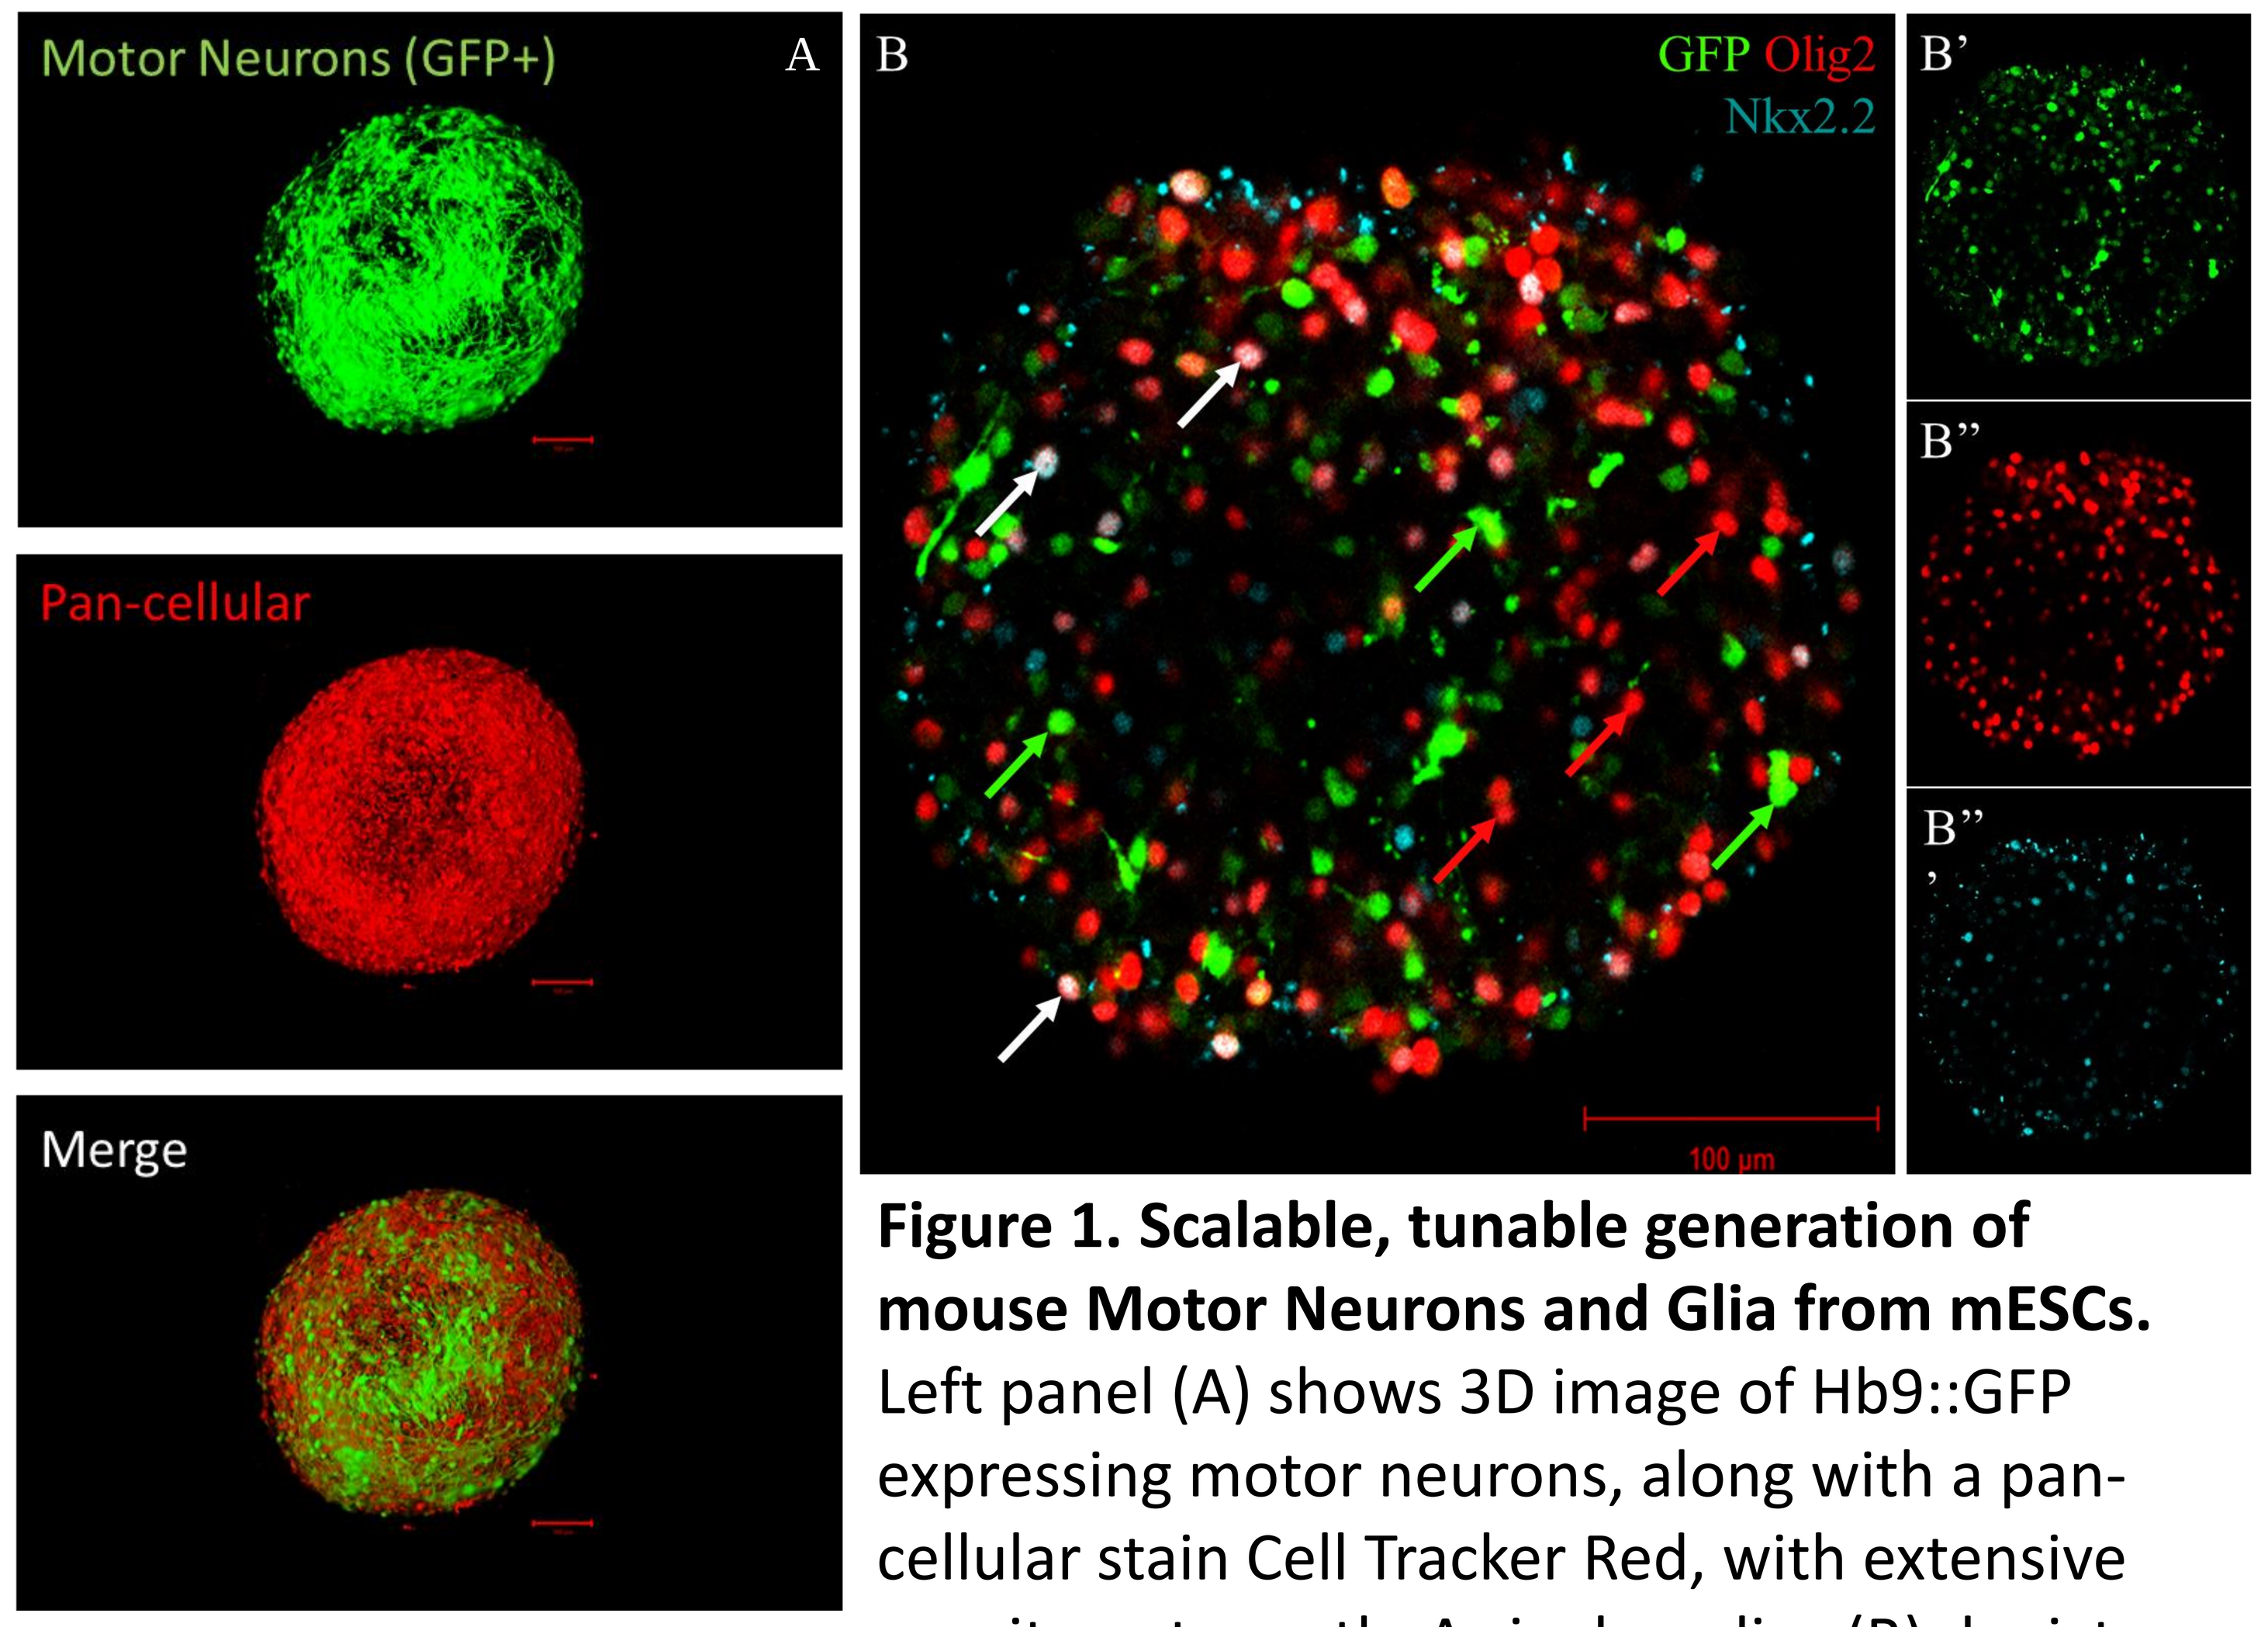
Motor Neurons (GFP+)
A
Pan-cellular
Merge
B
GFP Olig2
Nkx2.2
B’
B”
B”’
100 µm

Figure 1. Scalable, tunable generation of mouse Motor Neurons and Glia from mESCs. Left panel (A) shows 3D image of Hb9::GFP expressing motor neurons, along with a pan-cellular stain Cell Tracker Red, with extensive neurite outgrowth. A single z-slice (B) depicts three labeled proteins: GFP (B’), Olig2 (B”) and Nkx2.2 (B’’’). MN were denoted with GFP+ (green arrows, B), pMN with Olig2+/Nkx2.2- (red arrows, B), and OPC with Olig2+/Nkx2.2+ (white arrows, B). Images and slices were rebuilt using ZEN Software package (Carl Zeiss).

Methods

MEA Plating and Cell Culture:

•Plates: 12-, 48-, and 96-well MEAs (Axion BioSystems, Inc)

•Surface Coatings: PEI (0.1%) in borate buffer and Laminin (20mg/mL)

•Cell Density: 20,000-80,000 mMNGFP+ Mouse Motor Neurons (ArunA Biomedical) per 5uL drop over the electrodes of the MEA wells.

•Change media every 2-3 days using mMNGFP+ Mouse Motor Neuron Media. Culture mMNGFP+ Mouse Motor Neurons for up 30 days.

•Perform Maestro® recordings and compound testing between days 7 and 30

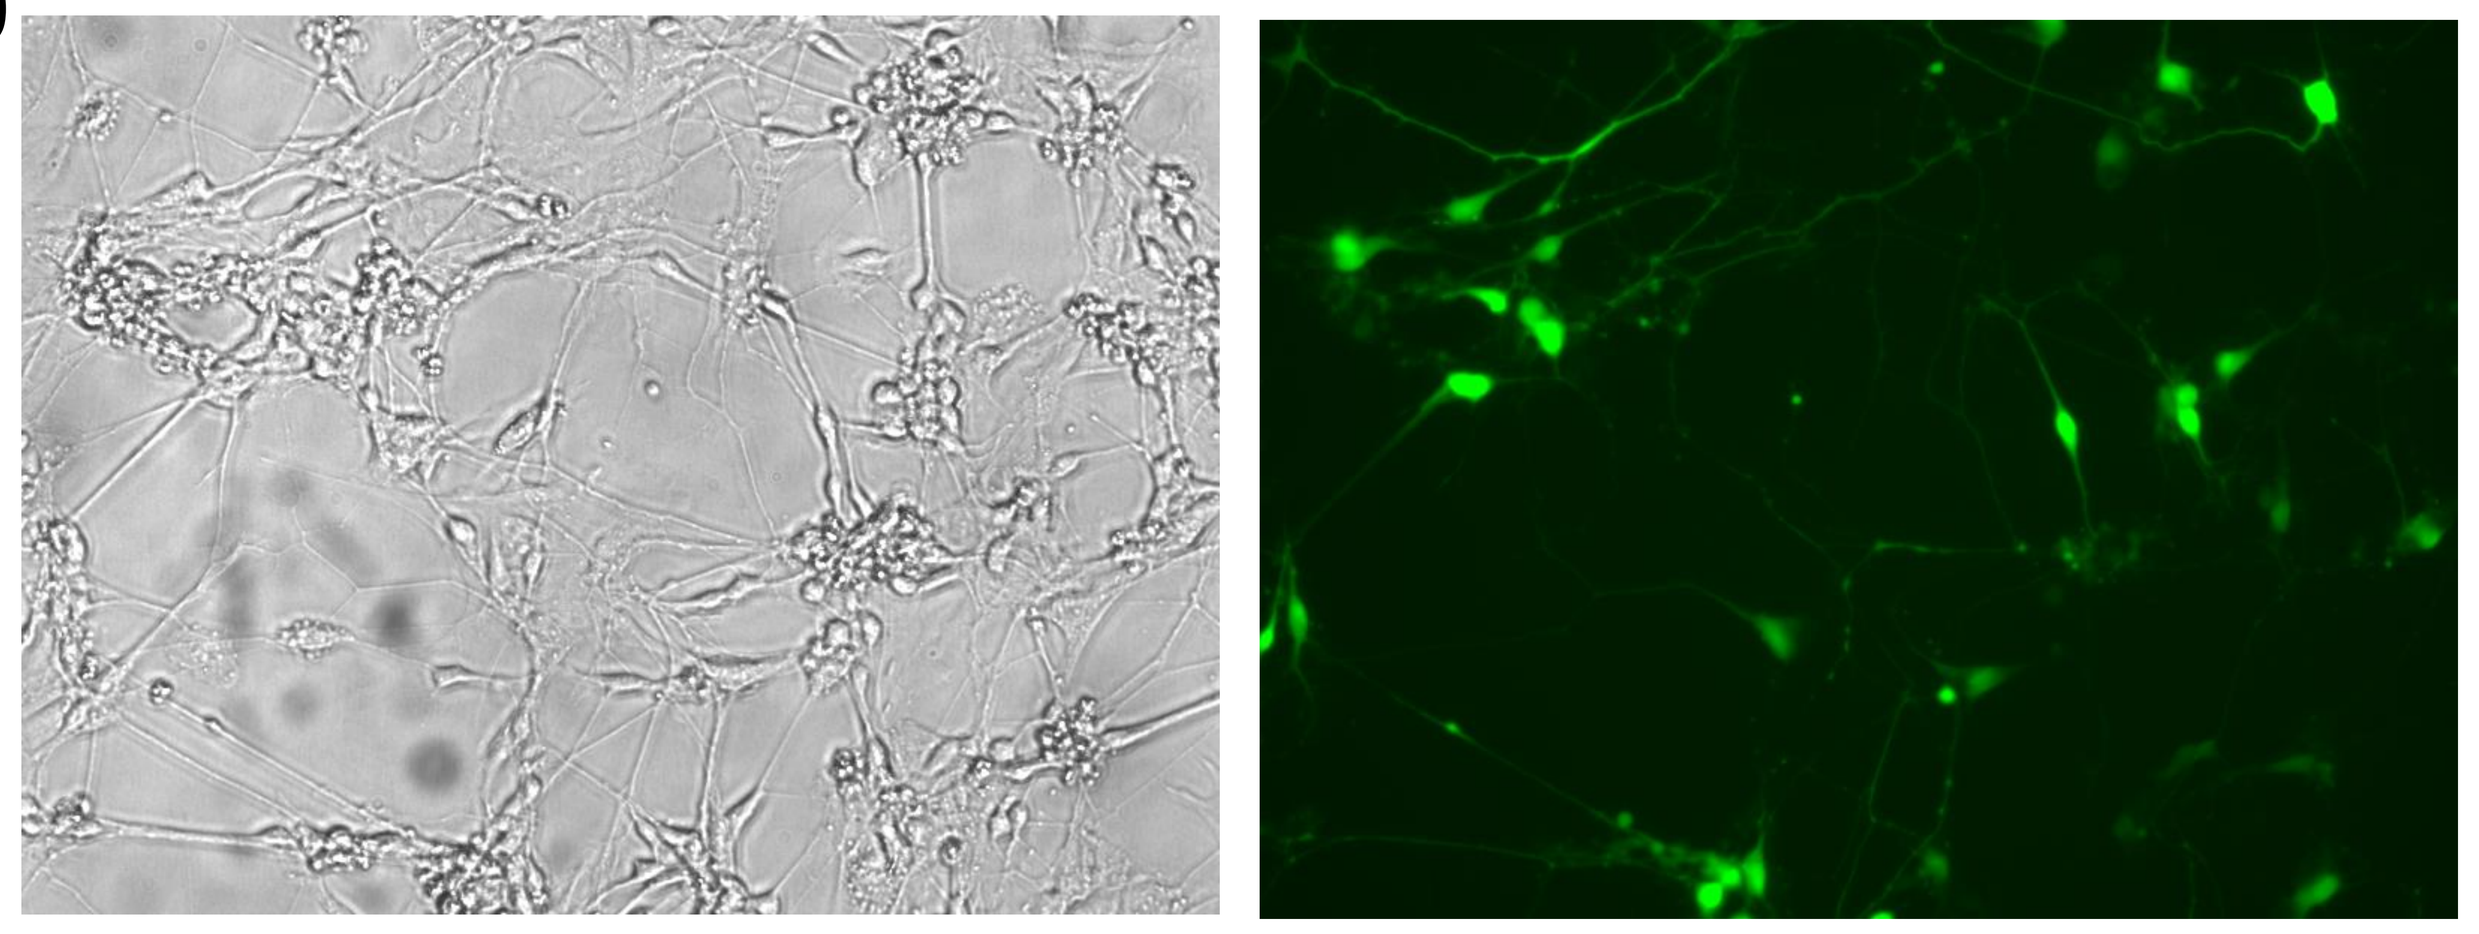

Figure 2. Post thaw phase and GFP mMN

Dosing Protocol:

Remove 50µL of conditioned media from each well of the MEA and transfer to a polystyrene plate. Prepare drug compounds (e.g. BIC) from stock and mix into the conditioned media in the polystyrene plate. Place MEA onto the Maestro and record a 5 minute baseline in Neural Spikes. Transfer 50µL of the conditioned media/drug compound mix back to the MEA, wait 15 minutes for drug equilibration, then record 5 minutes post dose.

Analysis:

Set the Spike Detector to 6X STD and generate a SPK file using AxIS, which contains the time and waveform specific metadata for each spike recorded.

Process the SPK file in Axion BioSystems’ Neural Metrics Tool to compile spike and burst statistics.

Summary

- Cryopreserved pre-differentiated motor neurons (GFP), interneurons (GABAergic) and glia derived from mouse ES can be thawed directly on MEA for quick assay implementation.
- Shortened time spike activity and bursting (starting at 6 DIV)
- Uniform genetic source reducing cell variability
- Optogenetics can be incorporated

Results

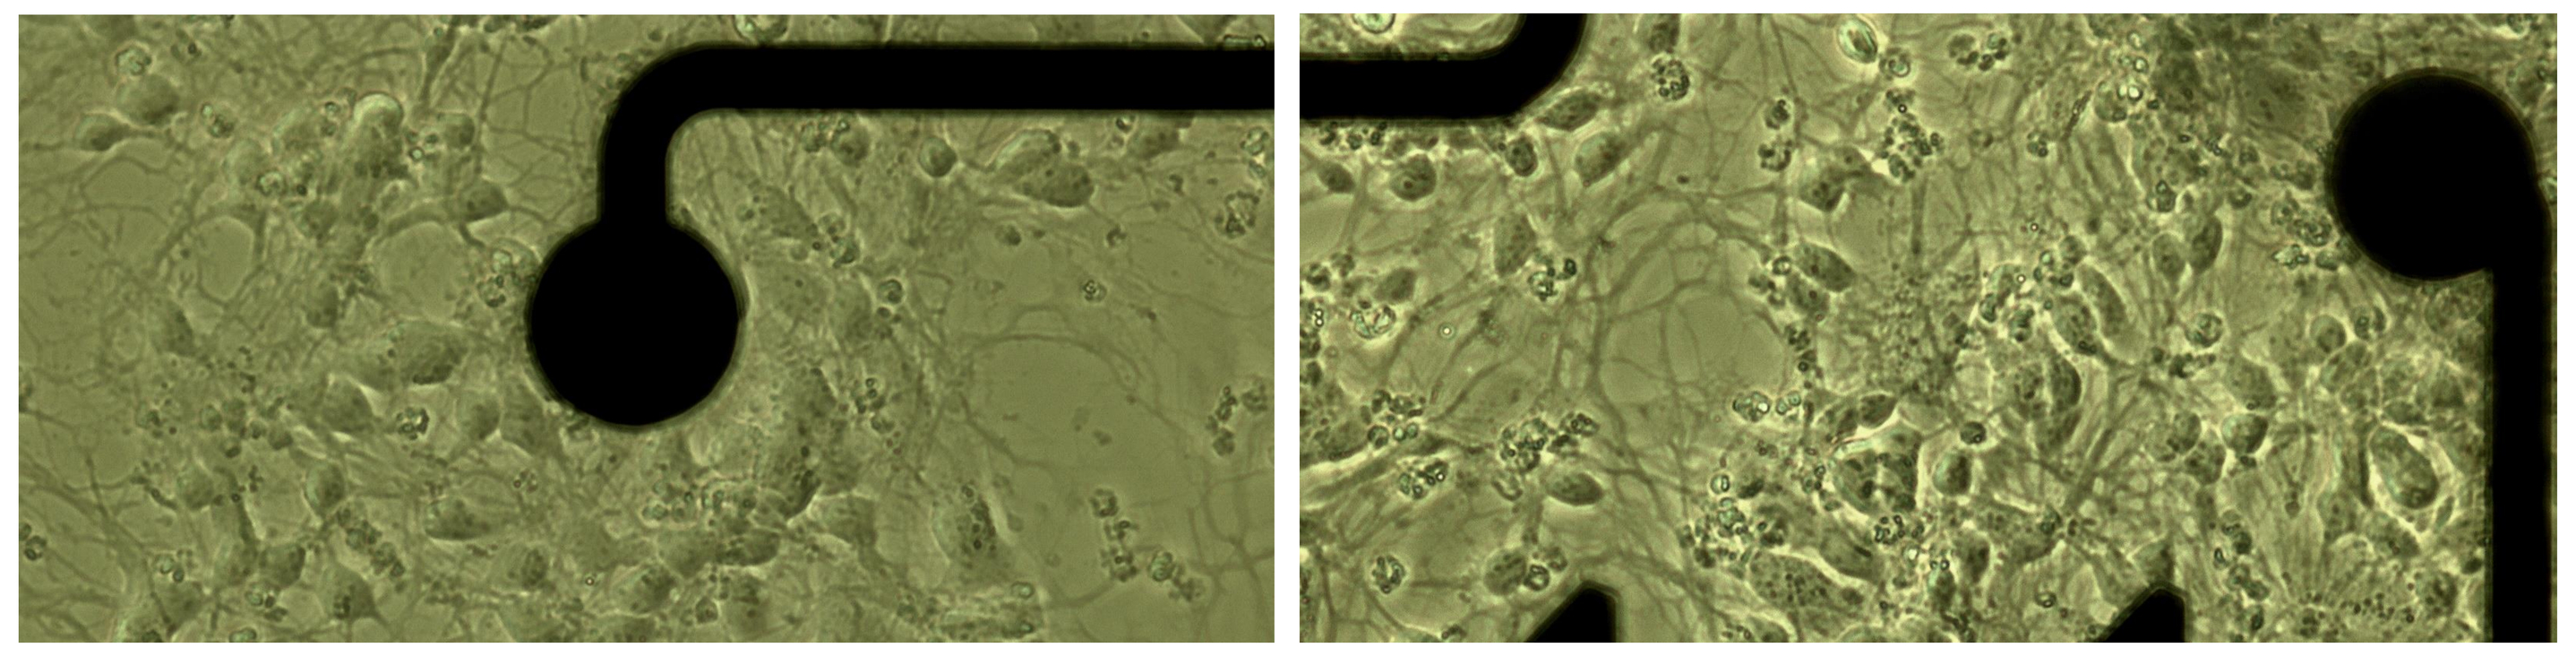

20K cells 2 DIV

60K cells 2 DIV

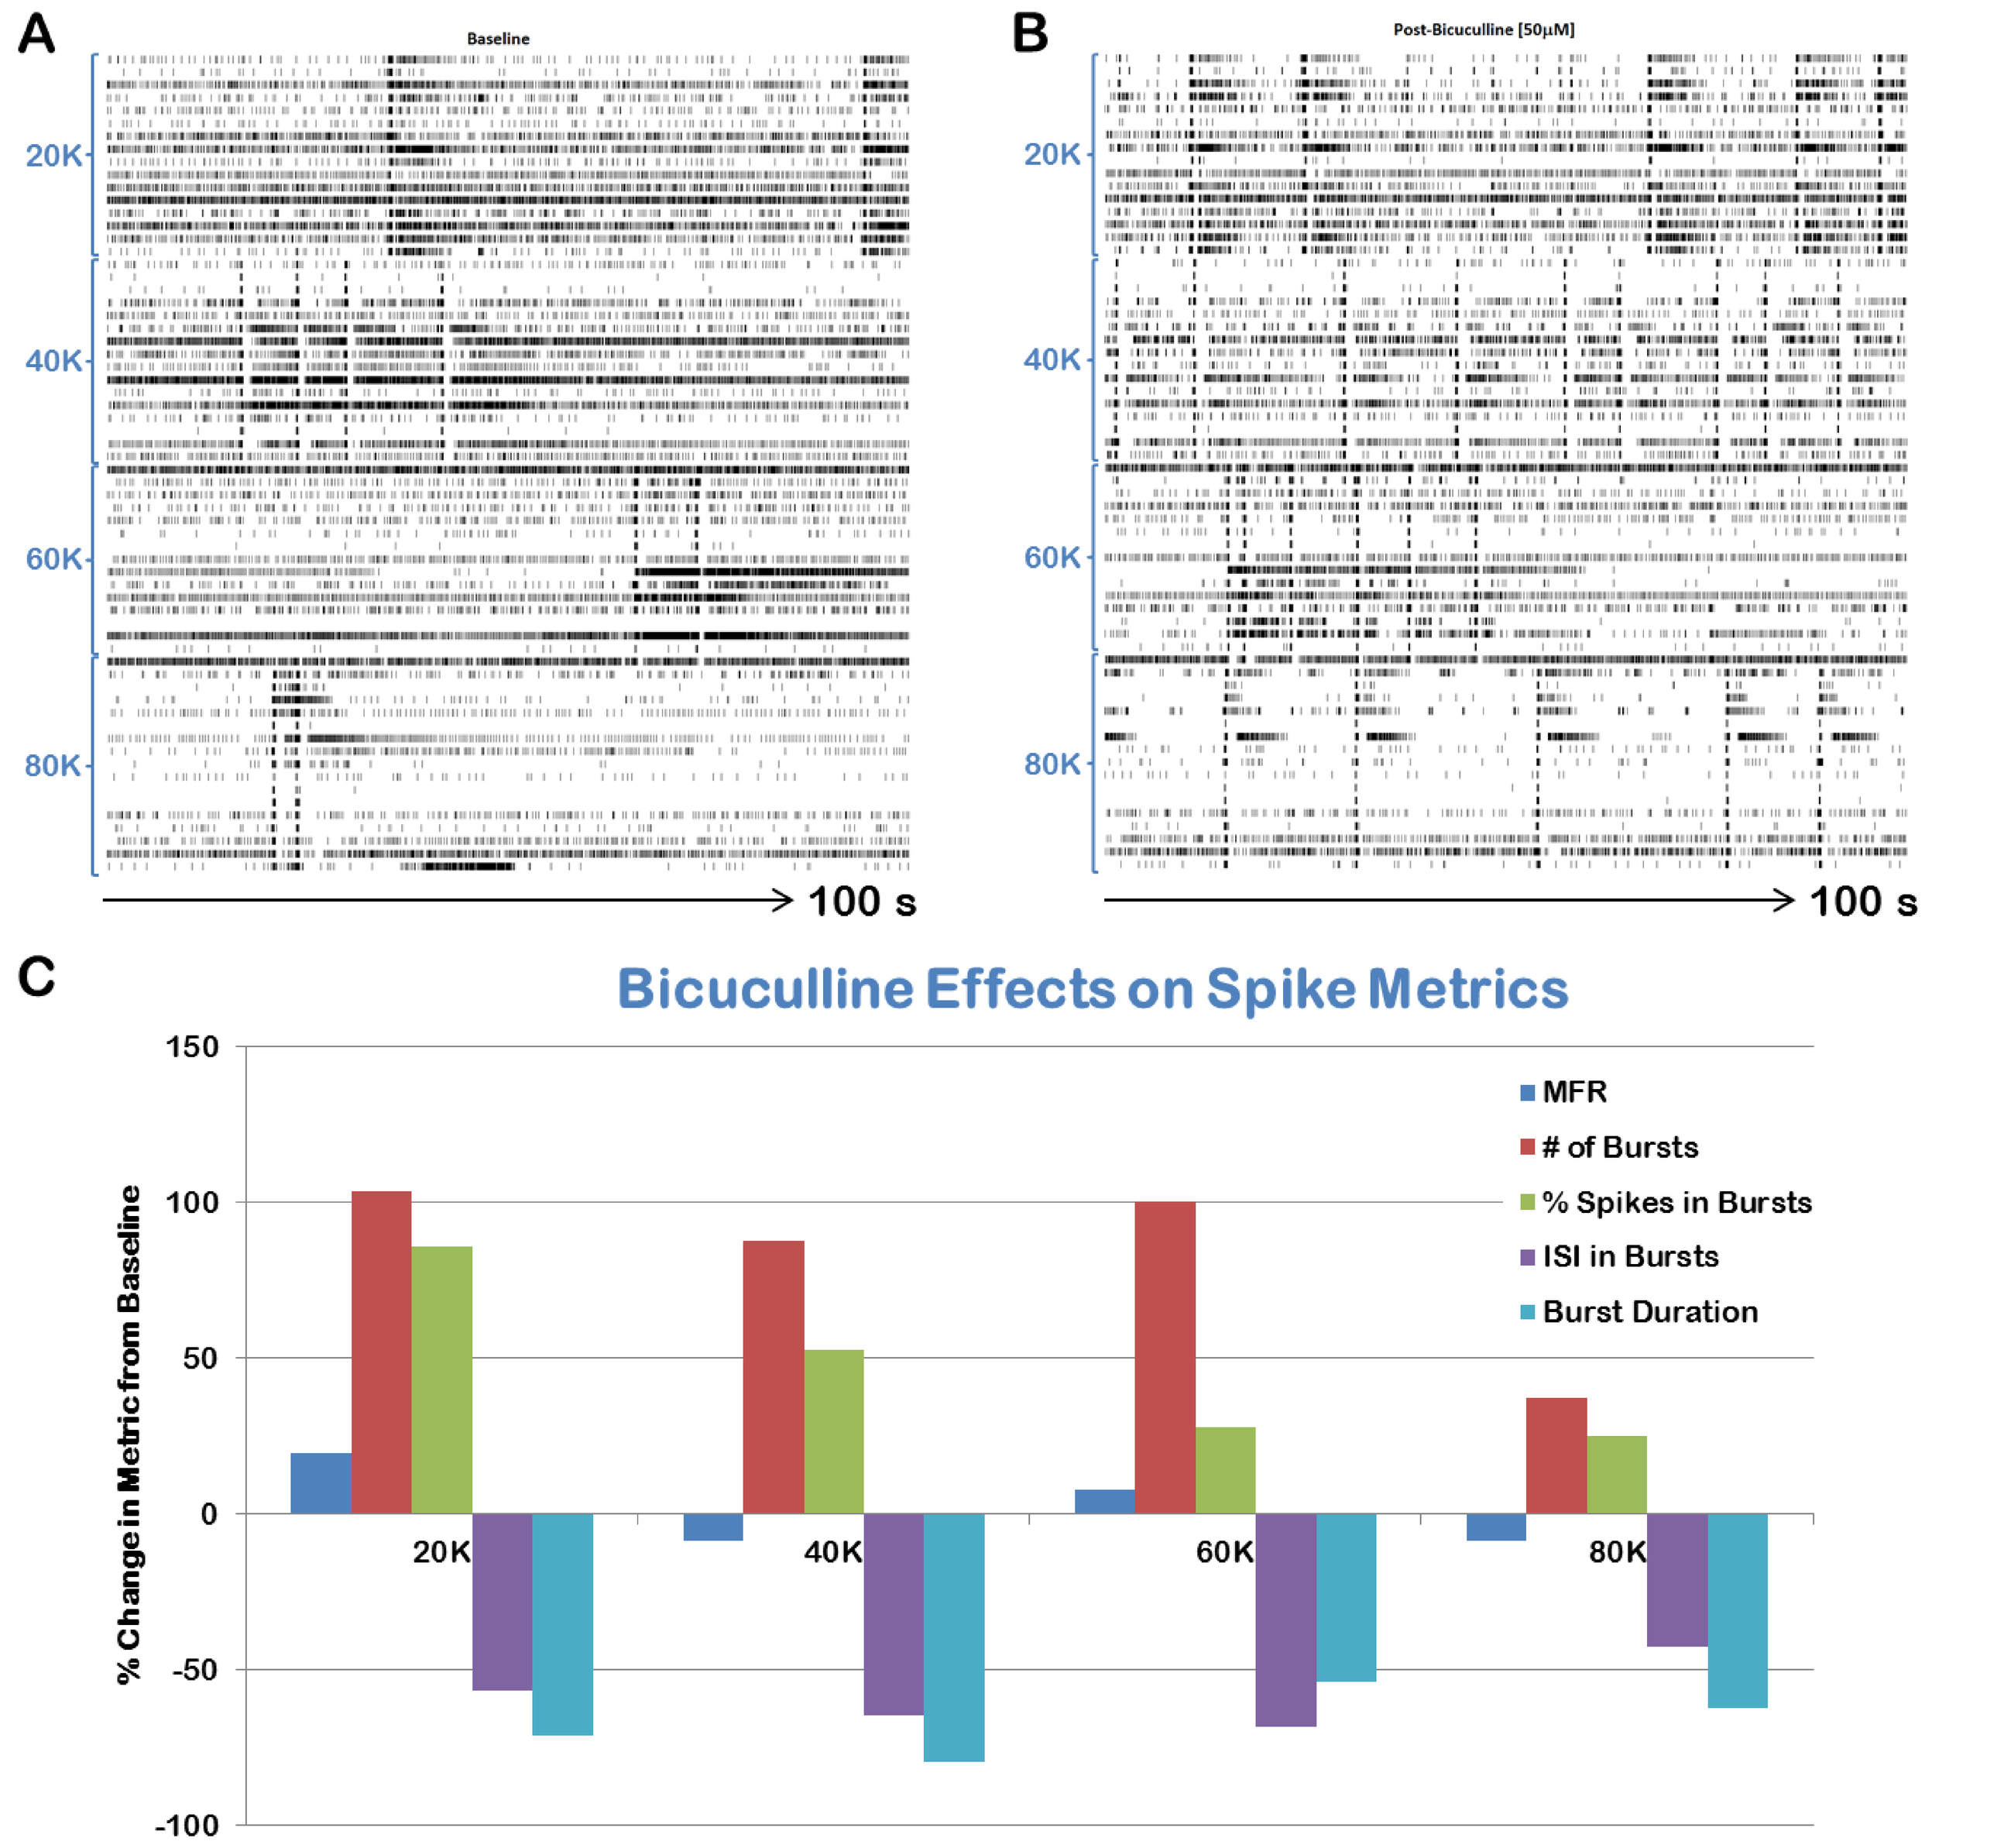
A
Baseline
B
Post-Bicuculline [50µM]
20K
40K
60K
80K
100 s
C
Bicuculline Effects on Spike Metrics
% Change in Metric from Baseline
150
100
50
0
-50
-100
20K
40K
60K
80K
MFR
of Bursts
% Spikes in Bursts
ISI in Bursts
Burst Duration

Figure 3: GABA-Antagonist Effects on ArunA mMNGFP+ Mouse Motor Neurons GFP+. Raster plots showing 100 second recordings from (A) baseline and (B)post-bicuculline treatment from 4 different cell densities of motor neurons. In response to bicuculline [50 µM], the burst activity increases and becomes more synchronized, as is evident from the rasters and in the % change of several metrics (C). Cell density increases demonstrate differences in several metrics including the number of bursts and the percentage of spikes detected in bursts.

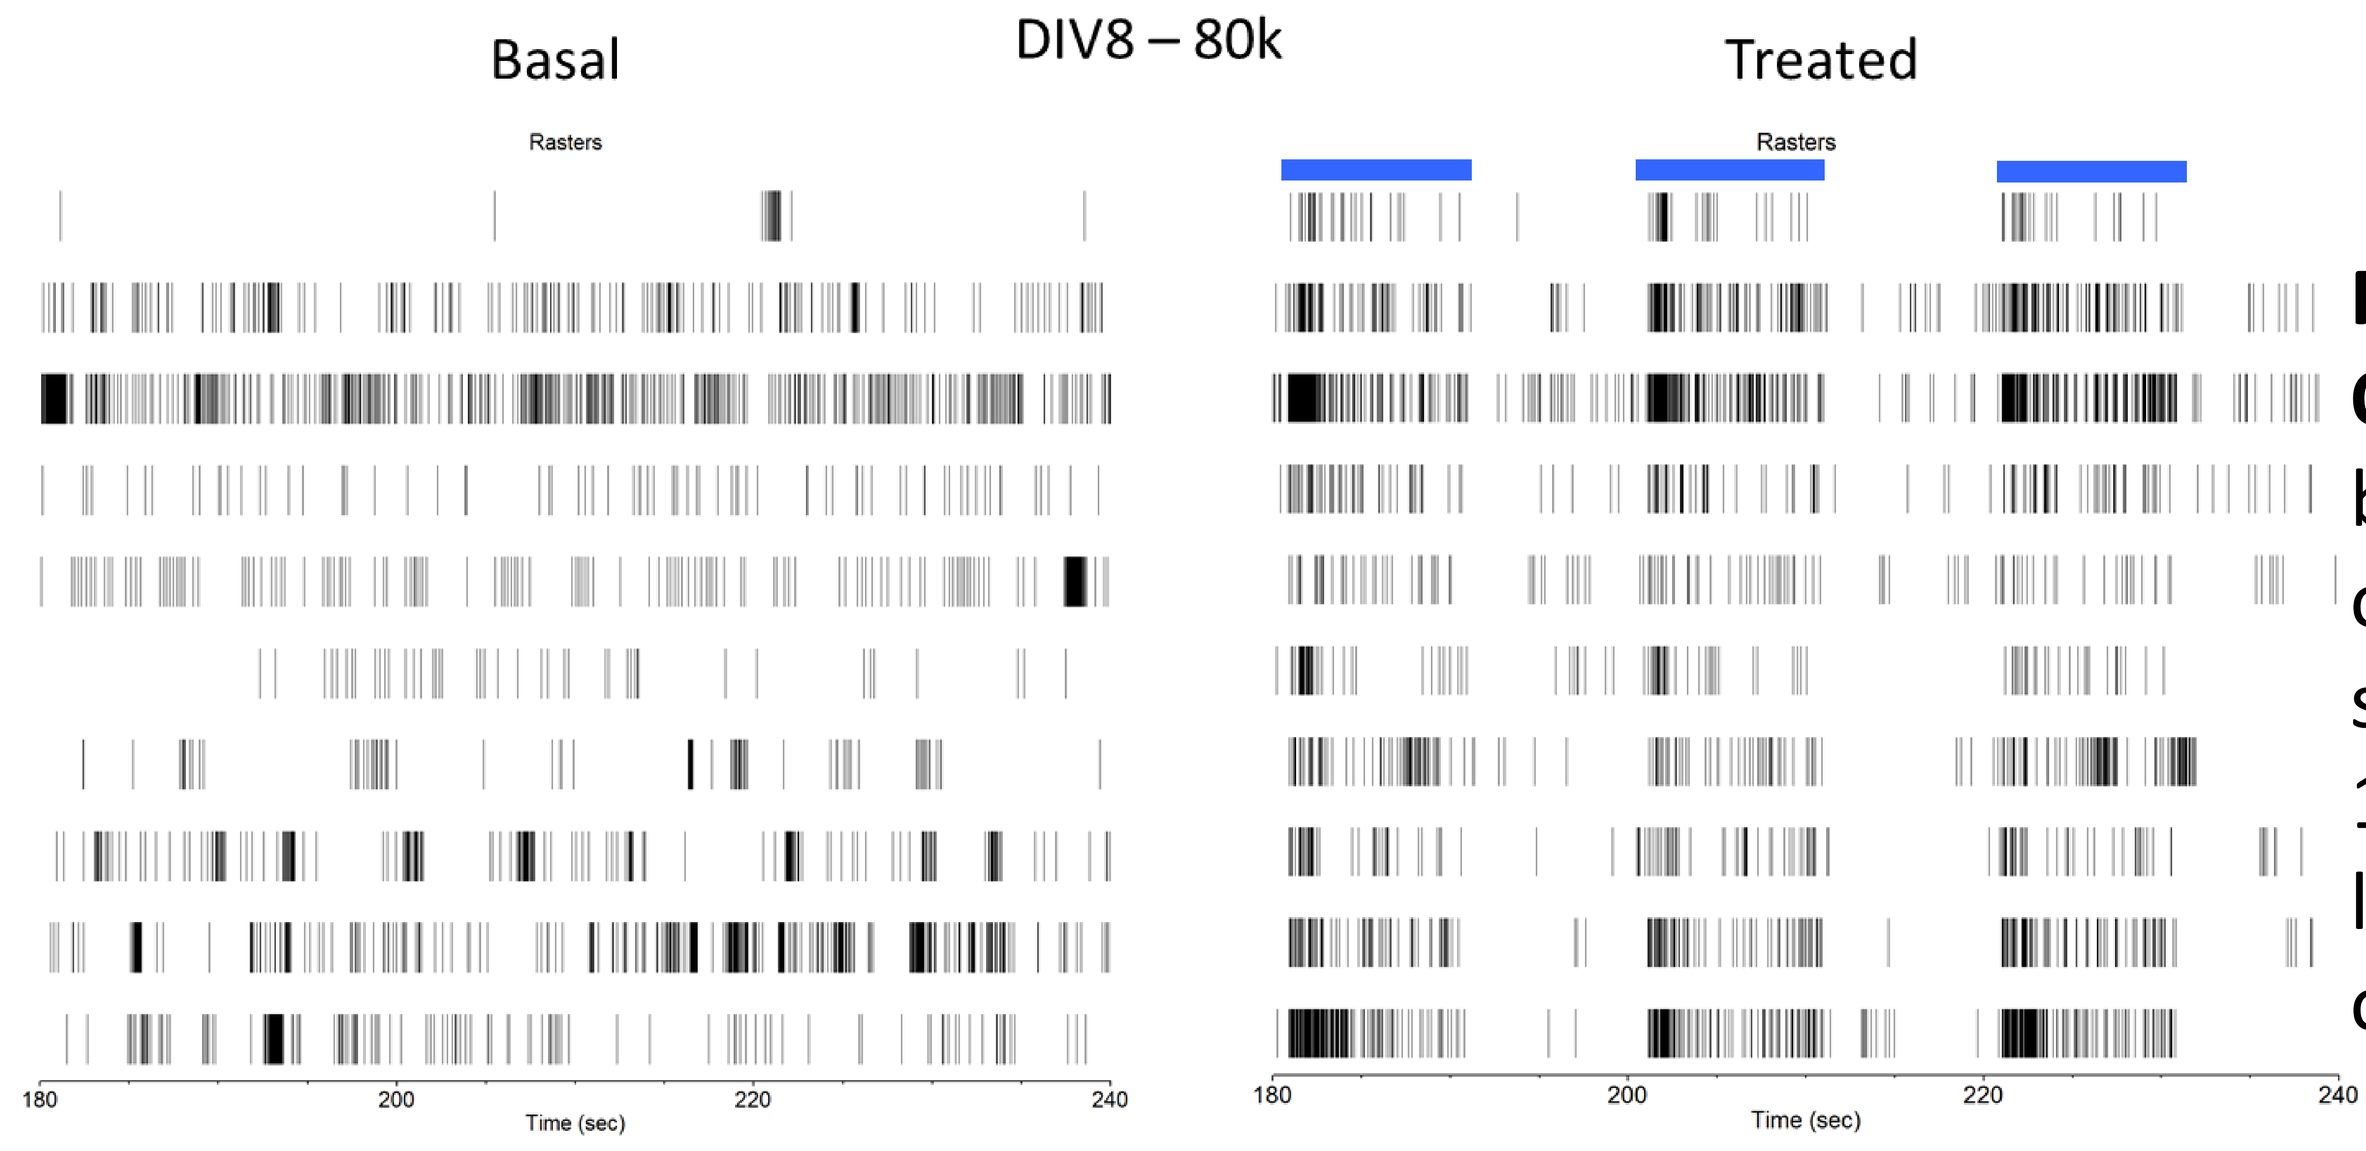
Basal
DIV8 – 80k
Treated
Rasters
180
200
220
240
Time (sec)

Figure 4. Optogenetics basal and optogenetic stimulation with 10 sec on (blue light) and 10 sec off repeated 3X